USING A WEBPORTAL TO ENHANCE NETWORK COMMUNICATION

Laurie McLeod, Stephanie Wellford, Emily Wilson
Rho, Inc.

Background

Rho serves as the coordinating center for several large, multi-center, collaborative research networks. Facilitating network communication can be challenging due to the number of people involved, varying schedules and time zones, and the volume of documents and versions distributed. In an effort to streamline the process, Rho created a secure network webportal to disseminate current information and materials to all network members in an efficient manner.

A Network in Numbers		
10 attendees per meeting	700 webportal users	6 time zones
4 documents per meeting		20 calls per week

The webportal houses general information, study documents, documents in development and meeting materials for network members to reference. This allows Rho to:

- Manage study and meeting materials in real-time
- Easily direct network members to information or documents
- Ensure all members access the same version of materials
- Reduce volume and size of members' email inboxes
- Collaboratively develop abstracts and manuscripts

Webportal Features	
Homepage Dashboard General documents Meeting materials Study-specific documents	Network committees Calendar Document development Staff directory

Login / Permissions

The webportal is secured through a log-in system, which requires users to enter a username and password to access the content. Rho manages user permissions to guarantee users can view the information that relates to their role in the network. Permissions also can be used to allow for distribution of materials and information to an exclusive group.

The navigation pane, shown below, is customized for the core network team members and the site team members. The network team has access to all studies, documents, and committees while the clinical site team only has access to the studies in which they take part.

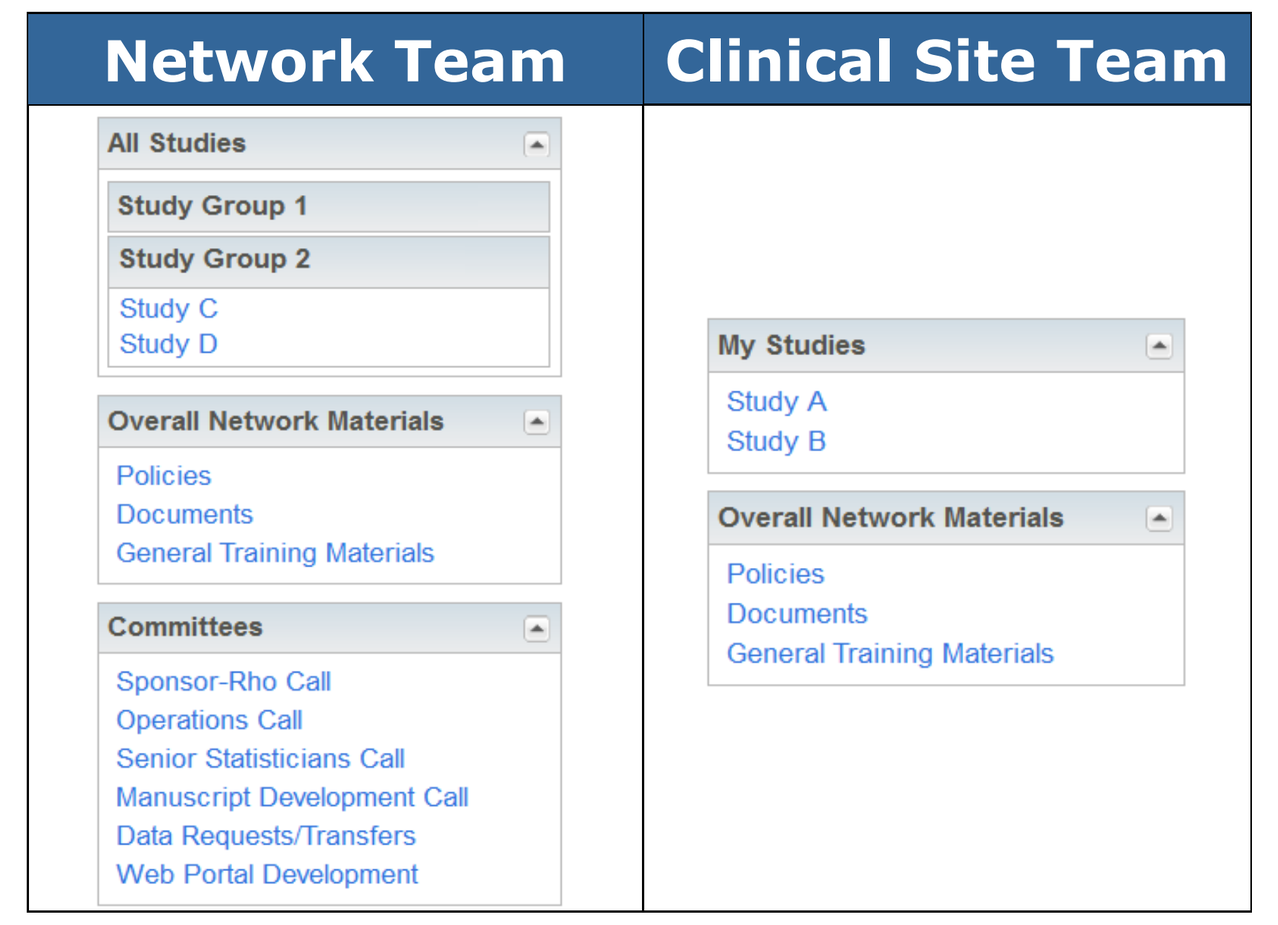
Network Team
Clinical Site Team
All Studies
Study Group 1
Study Group 2
Study C
Study D
Overall Network Materials
Policies
Documents
General Training Materials
Committees
Sponsor-Rho Call
Operations Call
Senior Statisticians Call
Manuscript Development Call
Data Requests/Transfers
Web Portal Development
My Studies
Study A
Study B
Overall Network Materials
Policies
Documents
General Training Materials

Homepage

The homepage serves as an index to the website and also presents information for each day. The navigation pane is found on the left side of the page. All network members can view the daily call information and announcements that apply to the entire network.

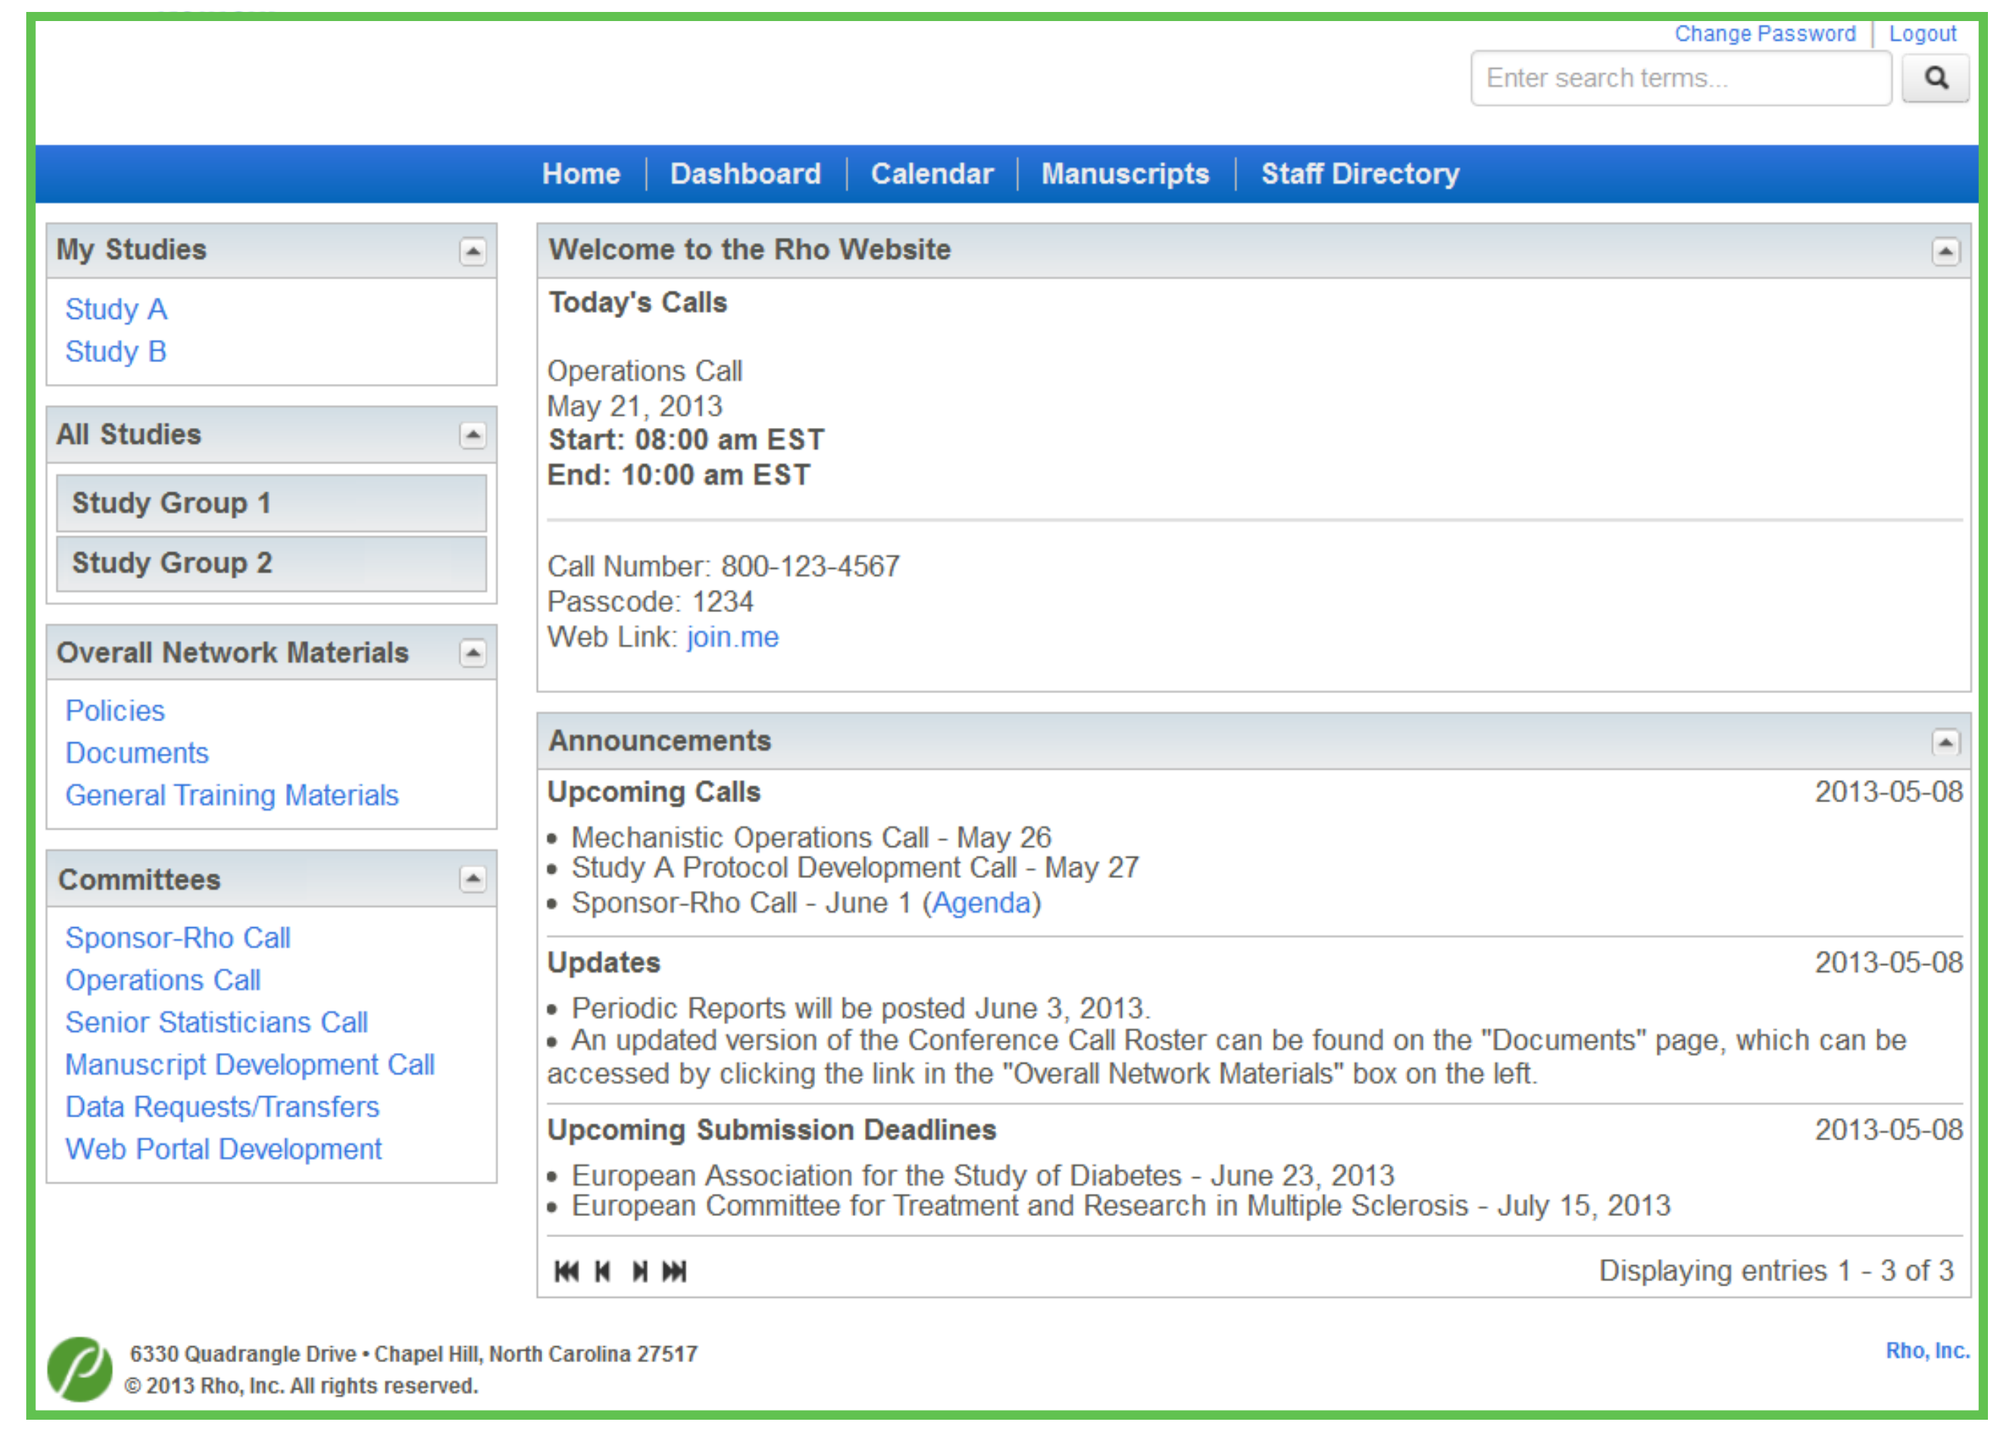
Change Password | Logout
Enter search terms...
Home | Dashboard | Calendar | Manuscripts | Staff Directory
My Studies
Study A
Study B
All Studies
Study Group 1
Study Group 2
Overall Network Materials
Policies
Documents
General Training Materials
Committees
Sponsor-Rho Call
Operations Call
Senior Statisticians Call
Manuscript Development Call
Data Requests/Transfers
Web Portal Development
Welcome to the Rho Website
Today's Calls
Operations Call
May 21, 2013
Start: 08:00 am EST
End: 10:00 am EST
Call Number: 800-123-4567
Passcode: 1234
Web Link: join.me
Announcements
Upcoming Calls 2013-05-08
Mechanistic Operations Call - May 26
Study A Protocol Development Call - May 27
Sponsor-Rho Call - June 1 (Agenda)
Updates 2013-05-08
Periodic Reports will be posted June 3, 2013.
An updated version of the Conference Call Roster can be found on the "Documents" page, which can be accessed by clicking the link in the "Overall Network Materials" box on the left.
Upcoming Submission Deadlines 2013-05-08
European Association for the Study of Diabetes - June 23, 2013
European Committee for Treatment and Research in Multiple Sclerosis - July 15, 2013
Displaying entries 1 - 3 of 3
6330 Quadrangle Drive • Chapel Hill, North Carolina 27517
© 2013 Rho, Inc. All rights reserved.
Rho, Inc.

Dashboard

The dashboard summarizes critical study information and displays it on a single page view. This enhanced view enables the user to comprehend a large amount of important information and compare and contrast study performance in a fraction of the time it would take if the information was dispersed across multiple pages. It provides overall network information but also allows the user to drill down into a particular study. This tool is available to all network members.

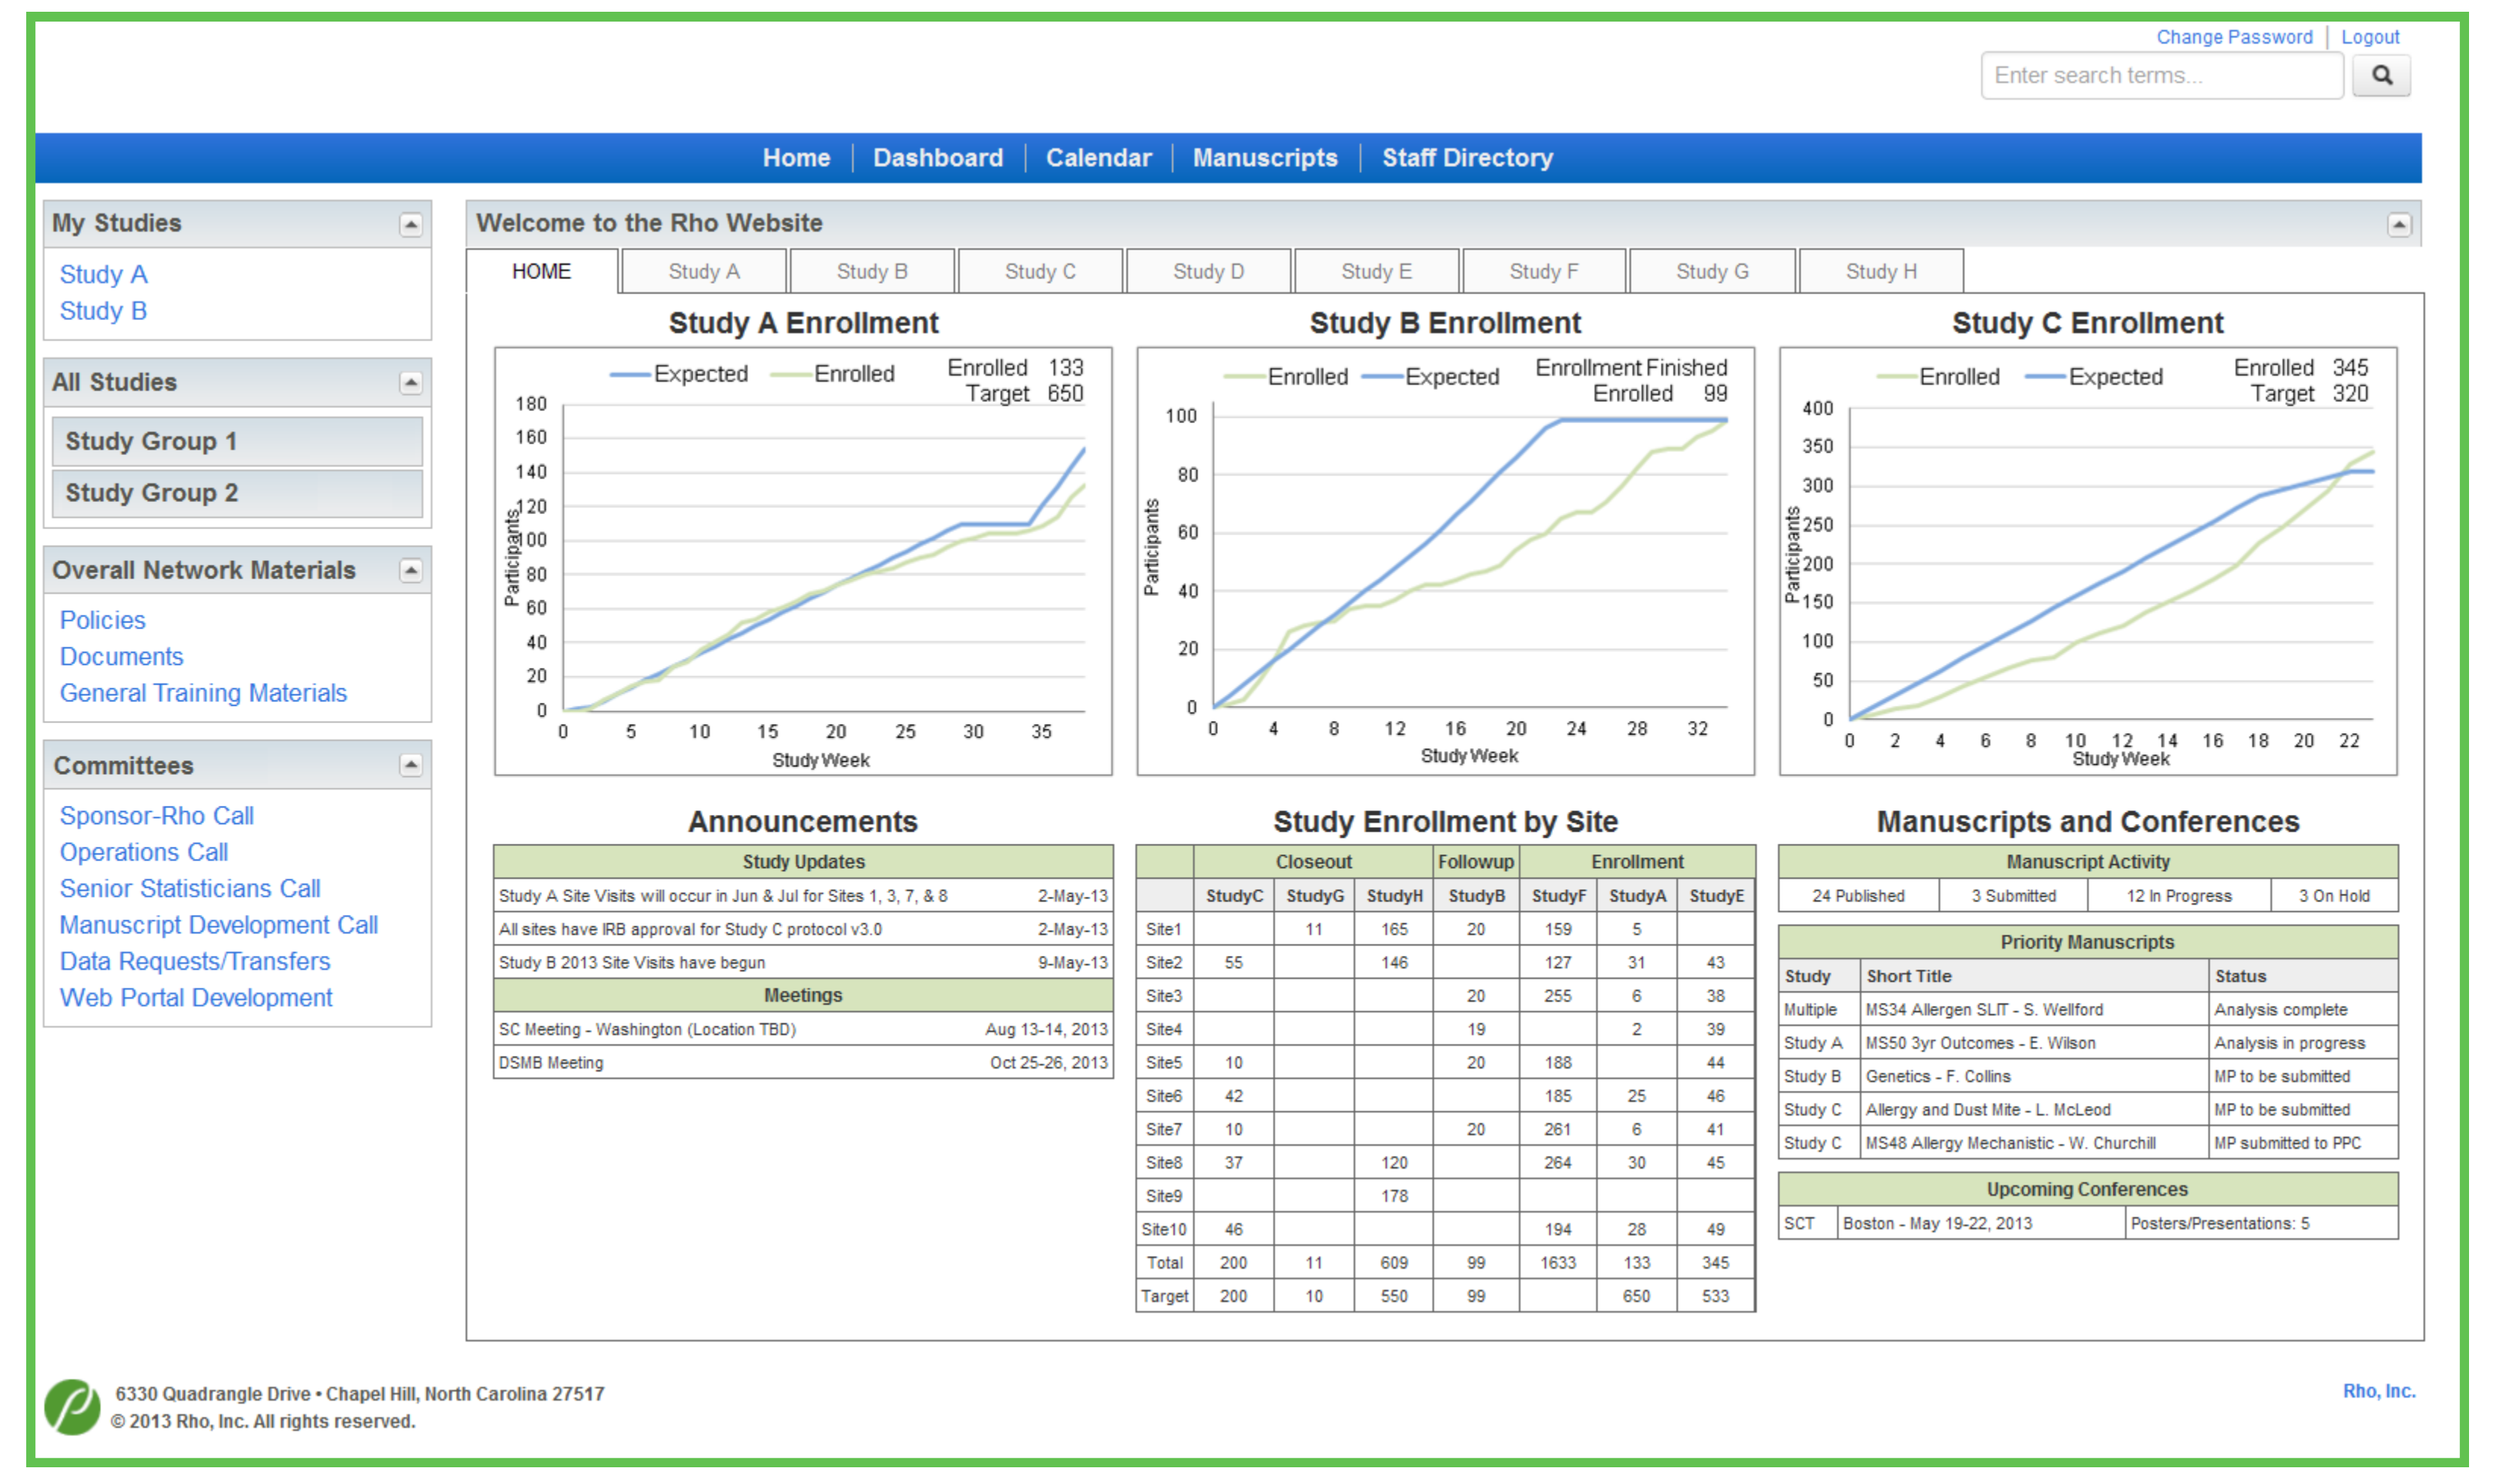
Change Password | Logout
Home | Dashboard | Calendar | Manuscripts | Staff Directory
Welcome to the Rho Website
HOME
Study A
Study B
Study C
Study D
Study E
Study F
Study G
Study H
Study A Enrollment
Study B Enrollment
Study C Enrollment
Expected
Enrolled
Participants
Study Week
Announcements
Study Updates
Meetings
Study Enrollment by Site
Manuscripts and Conferences
Manuscript Activity
Priority Manuscripts
Upcoming Conferences
Rho, Inc.

Resource Library

General resources and study documents are housed on the webportal to allow Rho to easily direct members to documents, and also to ensure network members reference the same version. All members have access to the general resources, but study-specific resources are tailored to each user's role.

General	Study-Specific
GCP/FDA guidance Network Manuals of Operations Regulatory forms/information Training tools Network policies Network procedural documents	Status reports Lab and Pharmacy guidelines Protocol documents IRB documents Trainings Manuals of Operations CRFs, handouts, and logs

Calendar

The network calendar stores all calls, meetings and any other information needed for network events during each month. Users can reference the calendar for times and passcodes for teleconferences or travel information for meetings. A link to call or meeting documents and references is embedded within each event listed in the calendar, which automatically directs network members to the appropriate study page.

Meeting Materials

Network members take part in many meetings and calls throughout the life of the network. Meeting agendas, meeting minutes, and reference documents are stored on the webportal to allow all meeting attendees to readily access current and past meeting information.

Some networks have in-person meetings for which travel arrangements are made and presentations are held. Travel details, meeting agendas, and slide sets are also maintained on the webportal.

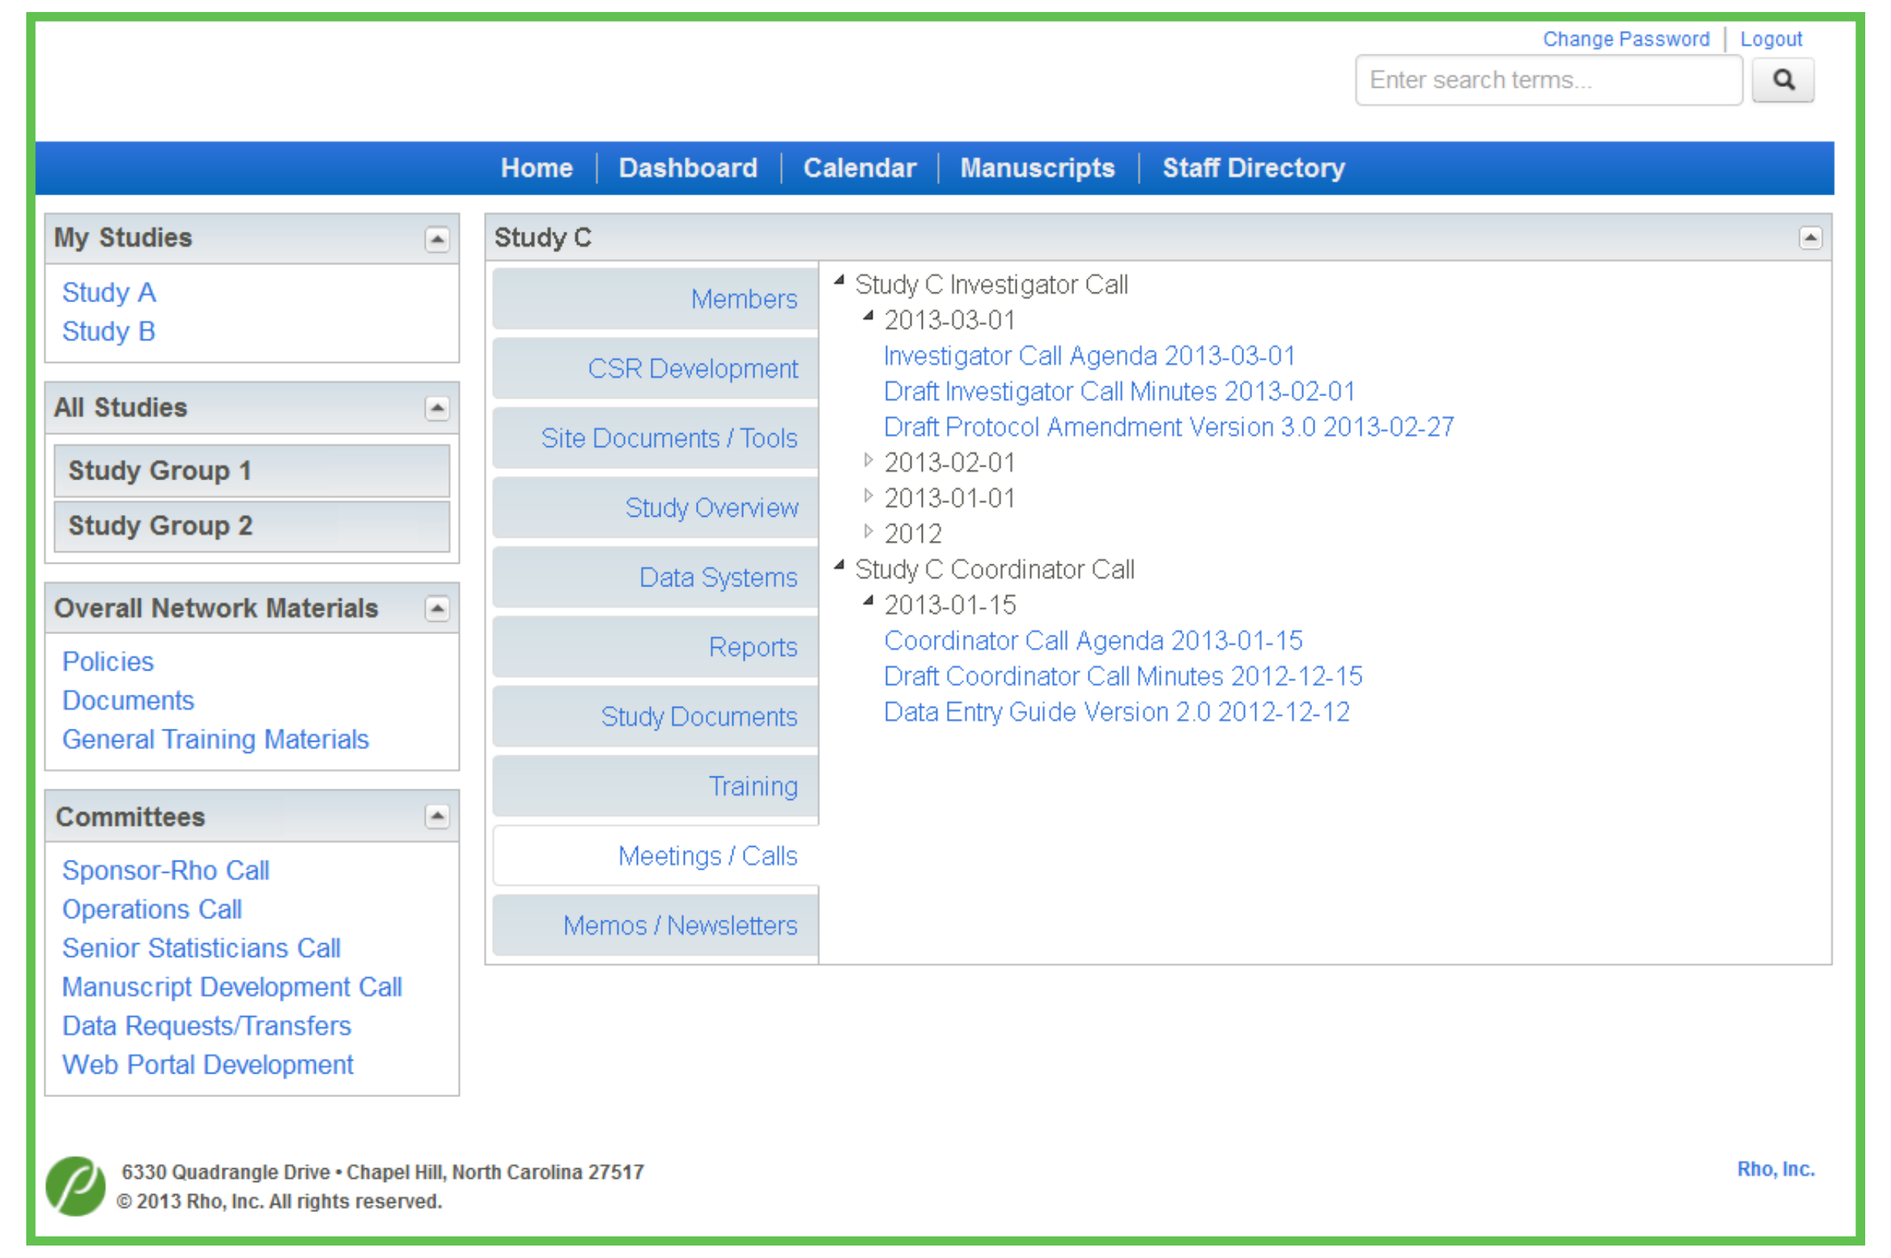
Change Password | Logout
Home | Dashboard | Calendar | Manuscripts | Staff Directory
Study C
Members
CSR Development
Site Documents / Tools
Study Overview
Data Systems
Reports
Study Documents
Training
Meetings / Calls
Memos / Newsletters
Study C Investigator Call
2013-03-01
Investigator Call Agenda 2013-03-01
Draft Investigator Call Minutes 2013-02-01
Draft Protocol Amendment Version 3.0 2013-02-27
2013-02-01
2013-01-01
2012
Study C Coordinator Call
2013-01-15
Coordinator Call Agenda 2013-01-15
Draft Coordinator Call Minutes 2012-12-15
Data Entry Guide Version 2.0 2012-12-12
Rho, Inc.

Document Development

The primary objective of clinical research is to gather information and share with the clinical research community. Collaborative abstracts, manuscripts, study reports, and other working documents are easily edited and re-posted through the document development tool, simplifying version control and streamlining the review process. This tool also creates an accessible, organized archive of all completed analyses and document versions for future reference.

Networks are often divided into committees that target specific tasks. Each committee has its own document development tool within its committee page, which enables committee members to edit, review, and upload comments and revisions.

In addition to regular document development, networks are often involved in various meetings at which data are presented and network progress is discussed. The webportal allows for presenters to post presentation materials for review or collaboration.

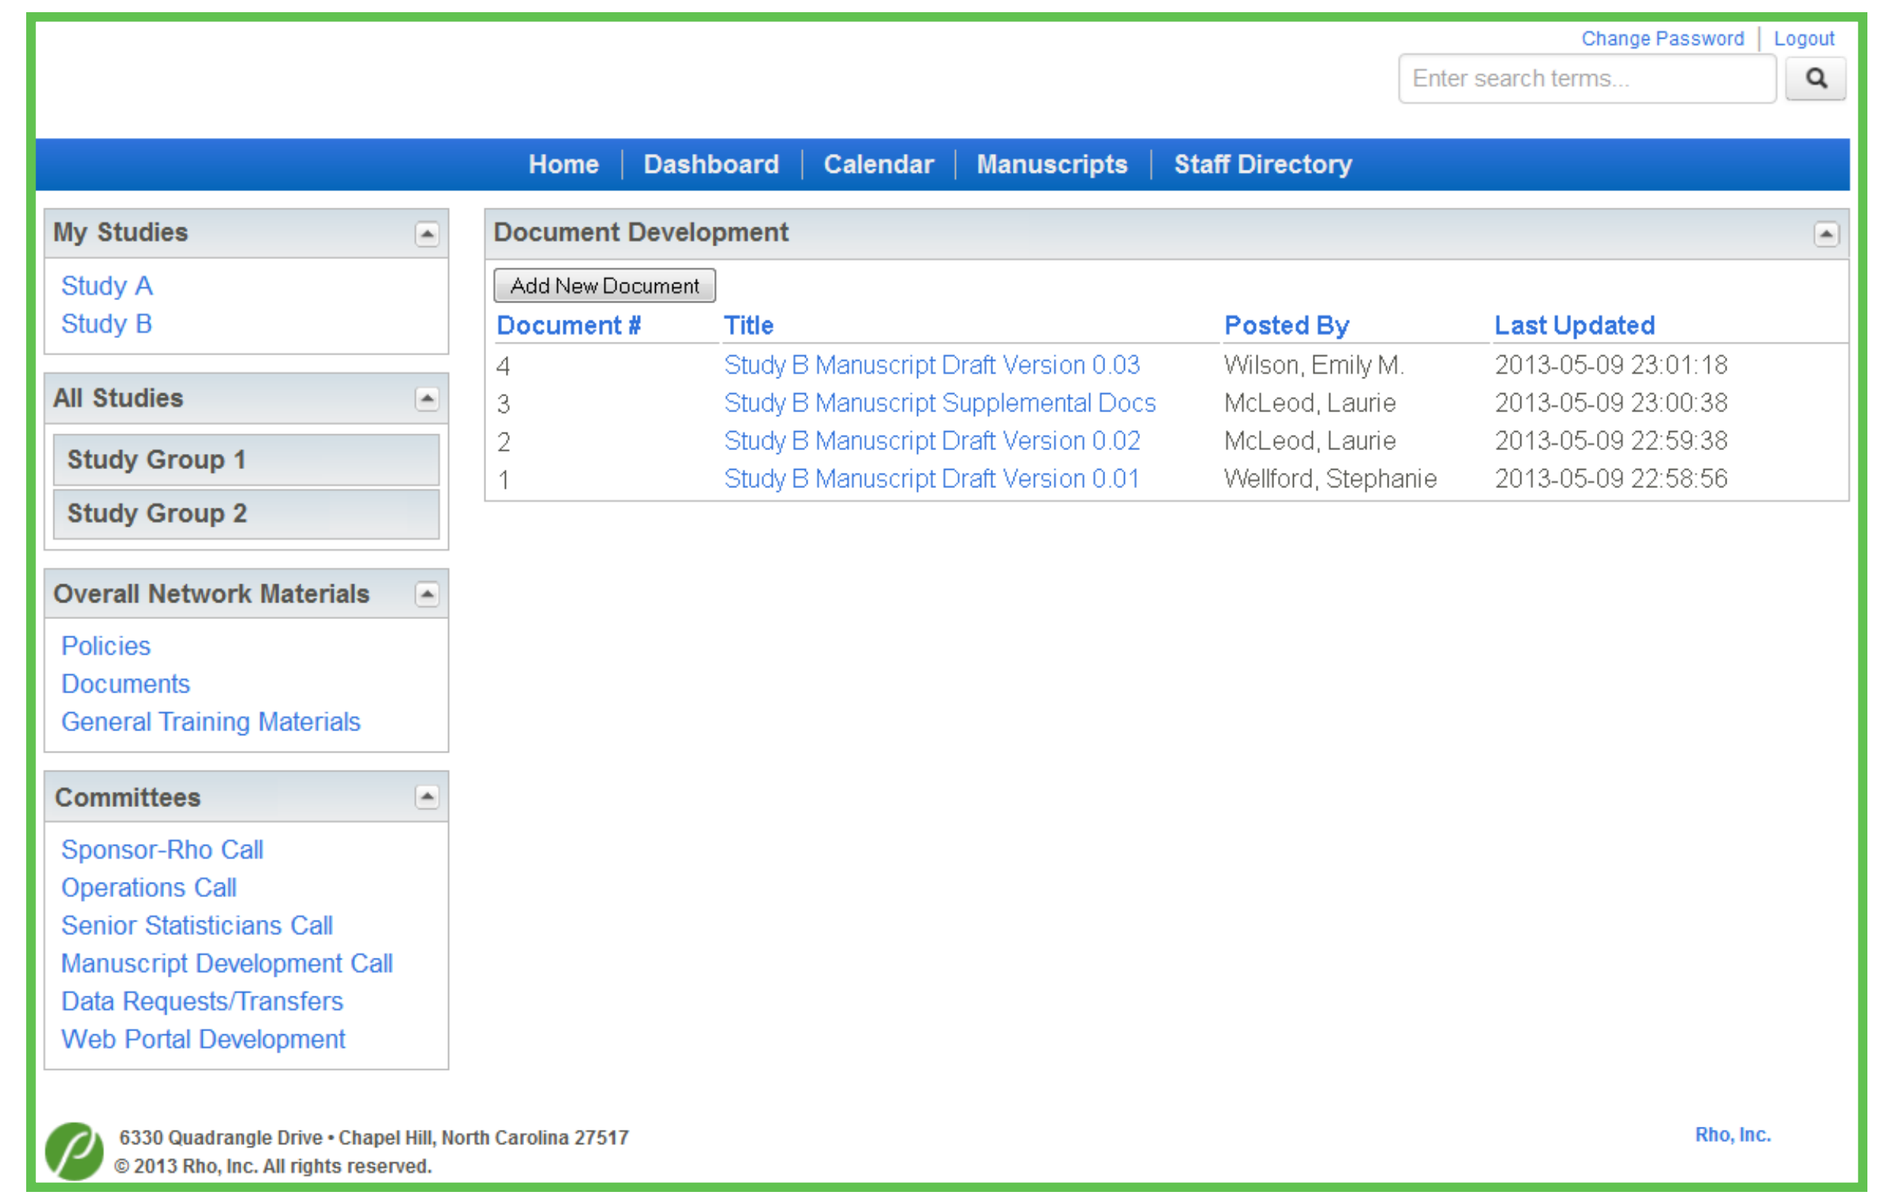
Change Password | Logout
Home | Dashboard | Calendar | Manuscripts | Staff Directory
Document Development
Add New Document
Document # | Title | Posted By | Last Updated
4 | Study B Manuscript Draft Version 0.03 | Wilson, Emily M. | 2013-05-09 23:01:18
3 | Study B Manuscript Supplemental Docs | McLeod, Laurie | 2013-05-09 23:00:38
2 | Study B Manuscript Draft Version 0.02 | McLeod, Laurie | 2013-05-09 22:59:38
1 | Study B Manuscript Draft Version 0.01 | Wellford, Stephanie | 2013-05-09 22:58:56
Rho, Inc.

Summary

The Rho webportal has proven to be a successful communication tool for our large networks. Its ability to house an unlimited number of documents allows for less clutter in all members' inboxes. The webportal also allows meetings and calls to be more efficient as Rho can direct committee members to a webpage rather than an email, also avoiding confusion by ensuring all network members are reviewing the most current version of the material. The unique resources, like the dashboard and document development system, eliminate the need to search through an array of emails, documents, and webpages to find the desired information. Since the webportal is readily available and accessible world-wide, network members across time zones and with varying schedules can access important network information anytime.

Rho®
Giving flight to research